Threshold Pricing Model: Not Just Another Cost-effectiveness Model

Deirdre Mladsi,[1] Stephanie Earnshaw,[1] Naoko Akashi-Ronquest,[1] Michael Keith[2]

[1] RTI Health Solutions, Research Triangle Park, NC, United States; [2] Shire Pharmaceuticals, Wayne, PA, United States

BACKGROUND

- Economic evaluation evolved, in part, to address payer concerns over pharmaceutical prices.
 - In the early days of economic evaluation, drugs produced net reductions in health care costs.
 - Manufacturers began reclaiming part of the societal benefit. Eventually, total costs of care began to increase with the introduction of new drugs.
 - Cost-effectiveness analysis (CEA) quantified the incremental cost required (net of all cost offsets) to get the resulting level of benefit.
- Payers began using CEA to inform pricing, reimbursement, and market access decisions. If the incremental cost associated with a new drug is deemed by a payer to be too much to pay to achieve the resulting health benefit, a payer may not be willing to pay the manufacturer's price, or the payer may restrict market access, for example, to a subset of the population most likely to benefit.
- Threshold analysis, as typically applied to cost-effectiveness models, is an extension of sensitivity analysis in which the threshold analysis may be used to demonstrate the maximum price opportunity that would result in the drug being considered cost-effective, given different levels of health outcome.
 - In traditional CEA, the incremental cost-effectiveness ratio (ICER) is the primary result of the model, and threshold analysis aids interpretation.
 - As a result, the model is restricted to the intended target indication, population, and line of therapy, reflecting key development decisions that have already been made.
- In contrast, a threshold pricing model, which is constructed early in a drug's development, does not attempt to evaluate the drug's cost-effectiveness. Instead, it informs the development plan, the pricing strategy, data gaps that need addressing, and go/no-go investment decisions.
- Given published and empirical evidence of a payer cost-effectiveness threshold, it is possible to anticipate and strategically plan to address pricing, reimbursement, and market access hurdles resulting from payer use of CEA. Specifically, it is possible for the manufacturer to estimate early in development the maximum value-based price opportunity associated with a new drug's target profile, prior to making key development decisions for the drug.
- The threshold value-based pricing model may reflect any number of scenarios reflecting various potential:
 - Indications
 - Positions in the treatment pathway
 - Comparators
 - Patient subgroups.
- Estimating and comparing value-based price opportunities for a new drug, prior to committing to the new drug's development plan, is a proxy for understanding the most important contribution to health that a new product can make.
- Table 1 presents the key differences between a traditional application of CEA and a threshold value-based pricing application of CEA.

Table 1. Traditional vs. Threshold Value-Based Pricing Application of CEA

Type of Difference	Traditional Application of CEA	Threshold Value-Based Pricing Application of CEA
Model output	Estimates ICER (e.g., for publication, health technology assessment, payer submission)	Estimates the following: • Value-based price opportunity, given an expected product profile • Minimum required efficacy and/or safety, given a target price • Individual contribution of various product attributes to value-based price
Timing	Conducted after drug price range has been narrowed (and after drug price has been used as the basis for investment decisions)	Conducted before setting target drug price and before investing in phase 3 trials
	Conducted after drug safety and efficacy are (nearly) known (near or at conclusion of phase 3 clinical trials)	Can clarify levels of safety and efficacy that would be required to support target price from a payer perspective
Perspective	Focuses on a particular indication, place in the treatment pathway, and comparator	Can evaluate the maximum value-based price opportunity over a wide range of potential indications, placements in the treatment pathway, subpopulations, and comparators
Audience	Provides little opportunity to influence product value for internal product decision making; rather it defines the product value for reimbursement and market access decision makers after characteristics of the product are known	Results can inform product profile, investment decisions, clinical trial design, and overall development plan for internal decision making

OBJECTIVE

- To outline the differences in the underlying mathematical structure, inputs, and outputs of a threshold pricing model compared with a traditional cost-effectiveness model, and to demonstrate its application.

METHODS

- Figure 1 presents the steps taken to develop a threshold pricing model, which are the same steps taken to develop a traditional cost-effectiveness model.

Figure 1. Steps for Threshold Pricing Model Development

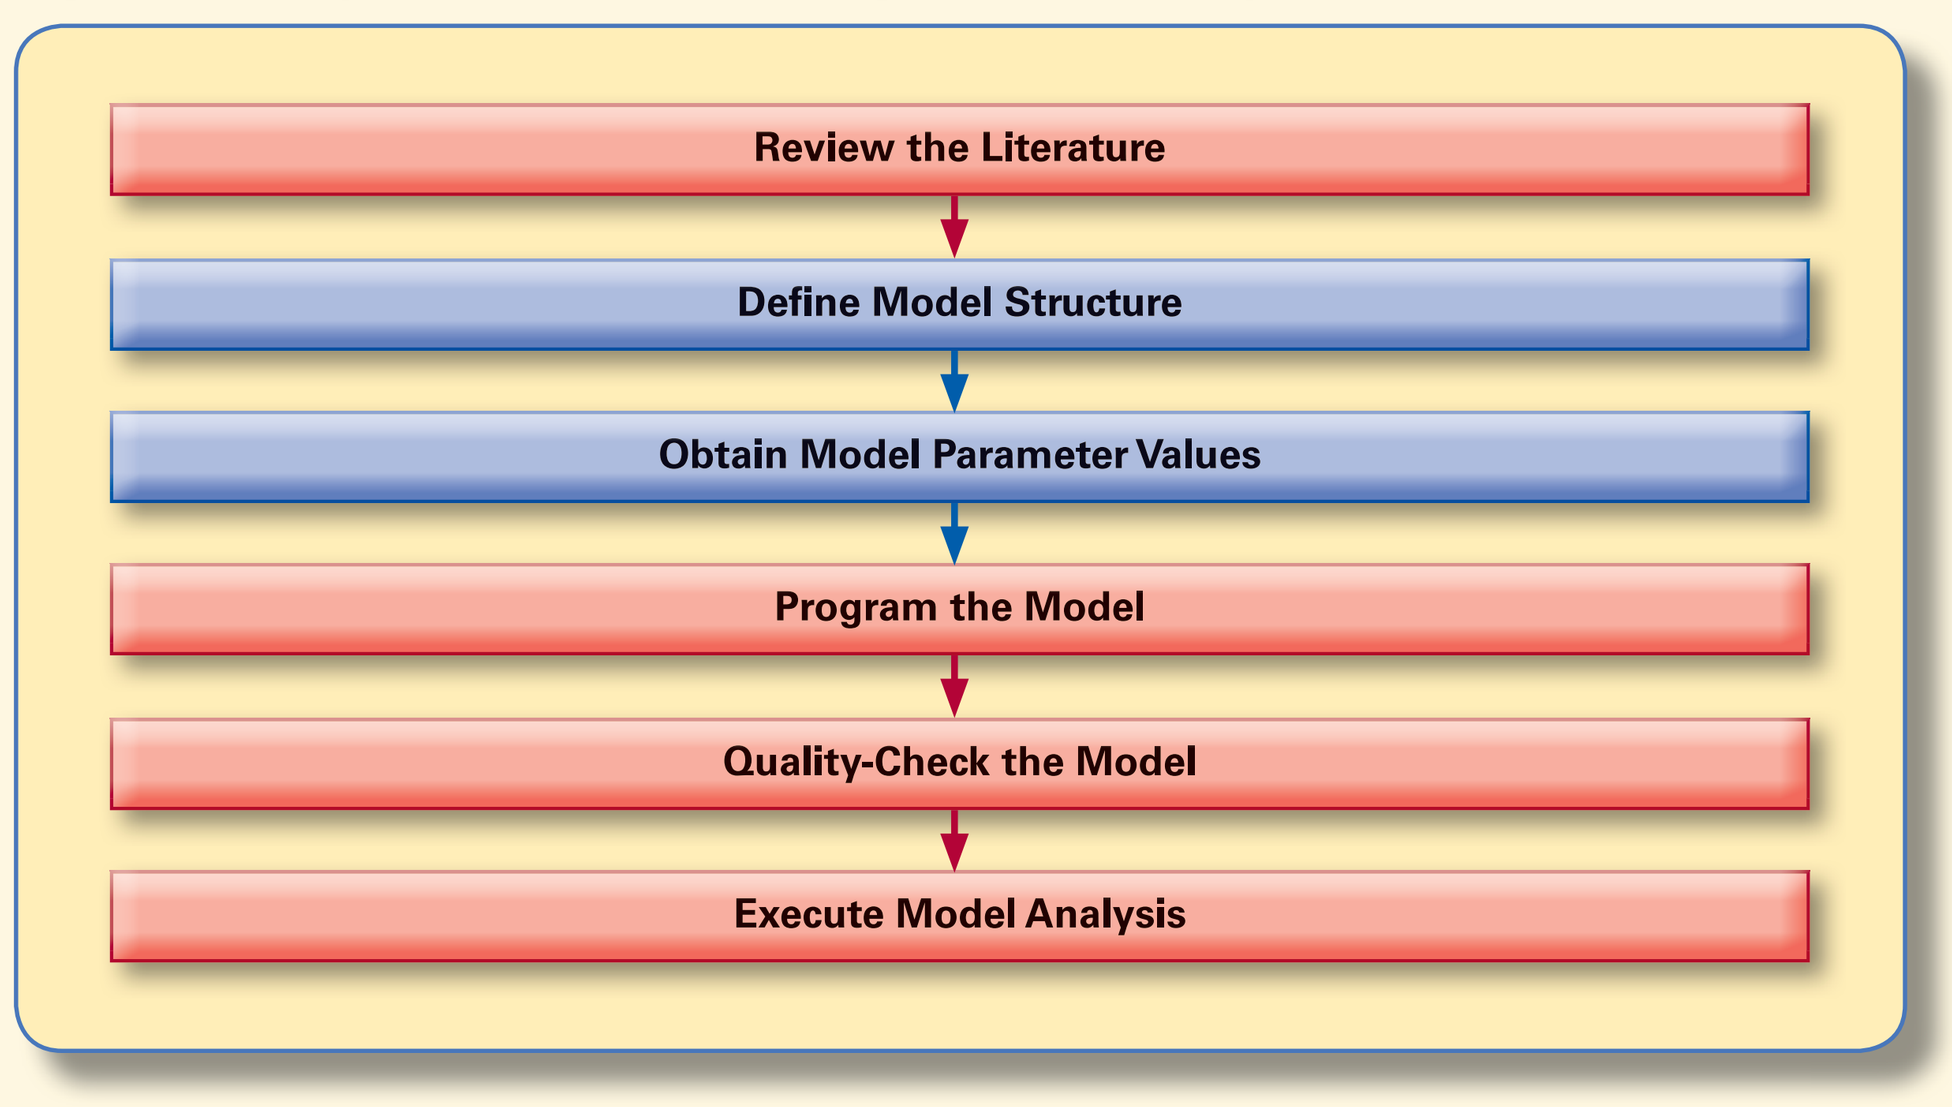

Review the Literature
Define Model Structure
Obtain Model Parameter Values
Program the Model
Quality-Check the Model
Execute Model Analysis

- A decision-analytic model developed for analyzing cost-effectiveness can be converted into a model to be used for estimating value-based price. We used algebraic manipulations to convert the ICER calculation into a value-based price calculation.
- Any number of decision-maker criteria (e.g., ICER, cost neutrality) may be used as the threshold in a threshold pricing model; however, typically the ICER is used (Equation 1).

Equation 1. ICER Equation

$$\boxed{ICER} = \frac{\Delta\,\text{Total Cost}_{(A\text{-}B)}}{\Delta\,\text{QALY}_{(A\text{-}B)}}$$

unknown

QALY = quality-adjusted life-year.

- Equation 2 depicts the ICER equation, but highlights the approach of solving the equation for new drug price or for new drug health benefit, given a known threshold ICER.

Equation 2. ICER Equation, Assuming Threshold ICER

unknown

$$ICER_T = \frac{\boxed{\text{Total Cost}_A} - \text{Total Cost}_B}{\boxed{\text{QALY}_A} - \text{QALY}_B}$$

unknown

- We used the following steps to solve for maximum value-based price opportunity for Drug A, given an expected level of efficacy of Drug A (which also may be accomplished using the Goal Seek function in Microsoft Excel):
 - Step 1: Solve for total treatment cost with Drug A that satisfies Equation 2 given the anticipated value of QALY A based on the target profile of Drug A.
 - Step 2: Solve for total drug cost for Drug A.
 - Step 3: Solve for unit price of Drug A.
- We used the following steps to solve for minimum required health benefit (e.g., efficacy, safety, tolerability, compliance, utility) of Drug A, given a target price for Drug A:
 - Step 1: Solve for total QALYs with Drug A that satisfies Equation 2 given the expected Total Cost A based on the target unit price for Drug A.
 - Step 2: Solve for efficacy of Drug A.
- We considered the case of a hypothetical new pharmaceutical that has not yet entered phase 3 (or perhaps even phase 2) clinical trials.
 - Based on the new pharmaceutical's mechanism of action, we assumed it would be similar to a currently marketed drug, but were confident that it would have a lower incidence of a key adverse event (AE), and that it might even have better efficacy and lower rates of other AEs.
 - We developed a target and an ideal version of the new drug's product profile, focusing on attributes that would be salient to a CEA (Table 2).
 - We anticipated pricing the drug at a 20% price premium to the currently marketed comparator with a similar mechanism of action (which was priced at £10 per day).

Table 2. Hypothetical New Pharmaceutical Target and Ideal Product Profiles

Attribute	Target	Ideal
Efficacy	Same as Drug B	X% better than Drug B
AE 1	X% better than Drug B	
AE 2	Similar to Drug B	
All other AEs	Similar to Drug B	

- Like the currently marketed product, the new pharmaceutical was expected to work in two related diseases.
- Also, it might assume a position in the treatment pathway for either disease as first-, second-, third-, or fourth-line treatment, each line of which would offer a set of potential comparators.
- Finally, there were multiple patient subgroups (e.g., based on age, comorbidities) that might be helped differentially by the new drug.
- Using methods equivalent to those that would be used to develop a traditional cost-effectiveness model, a threshold value-based pricing economic model structure for the two similar diseases was developed (Figure 2).

Figure 2. Economic Model Structure

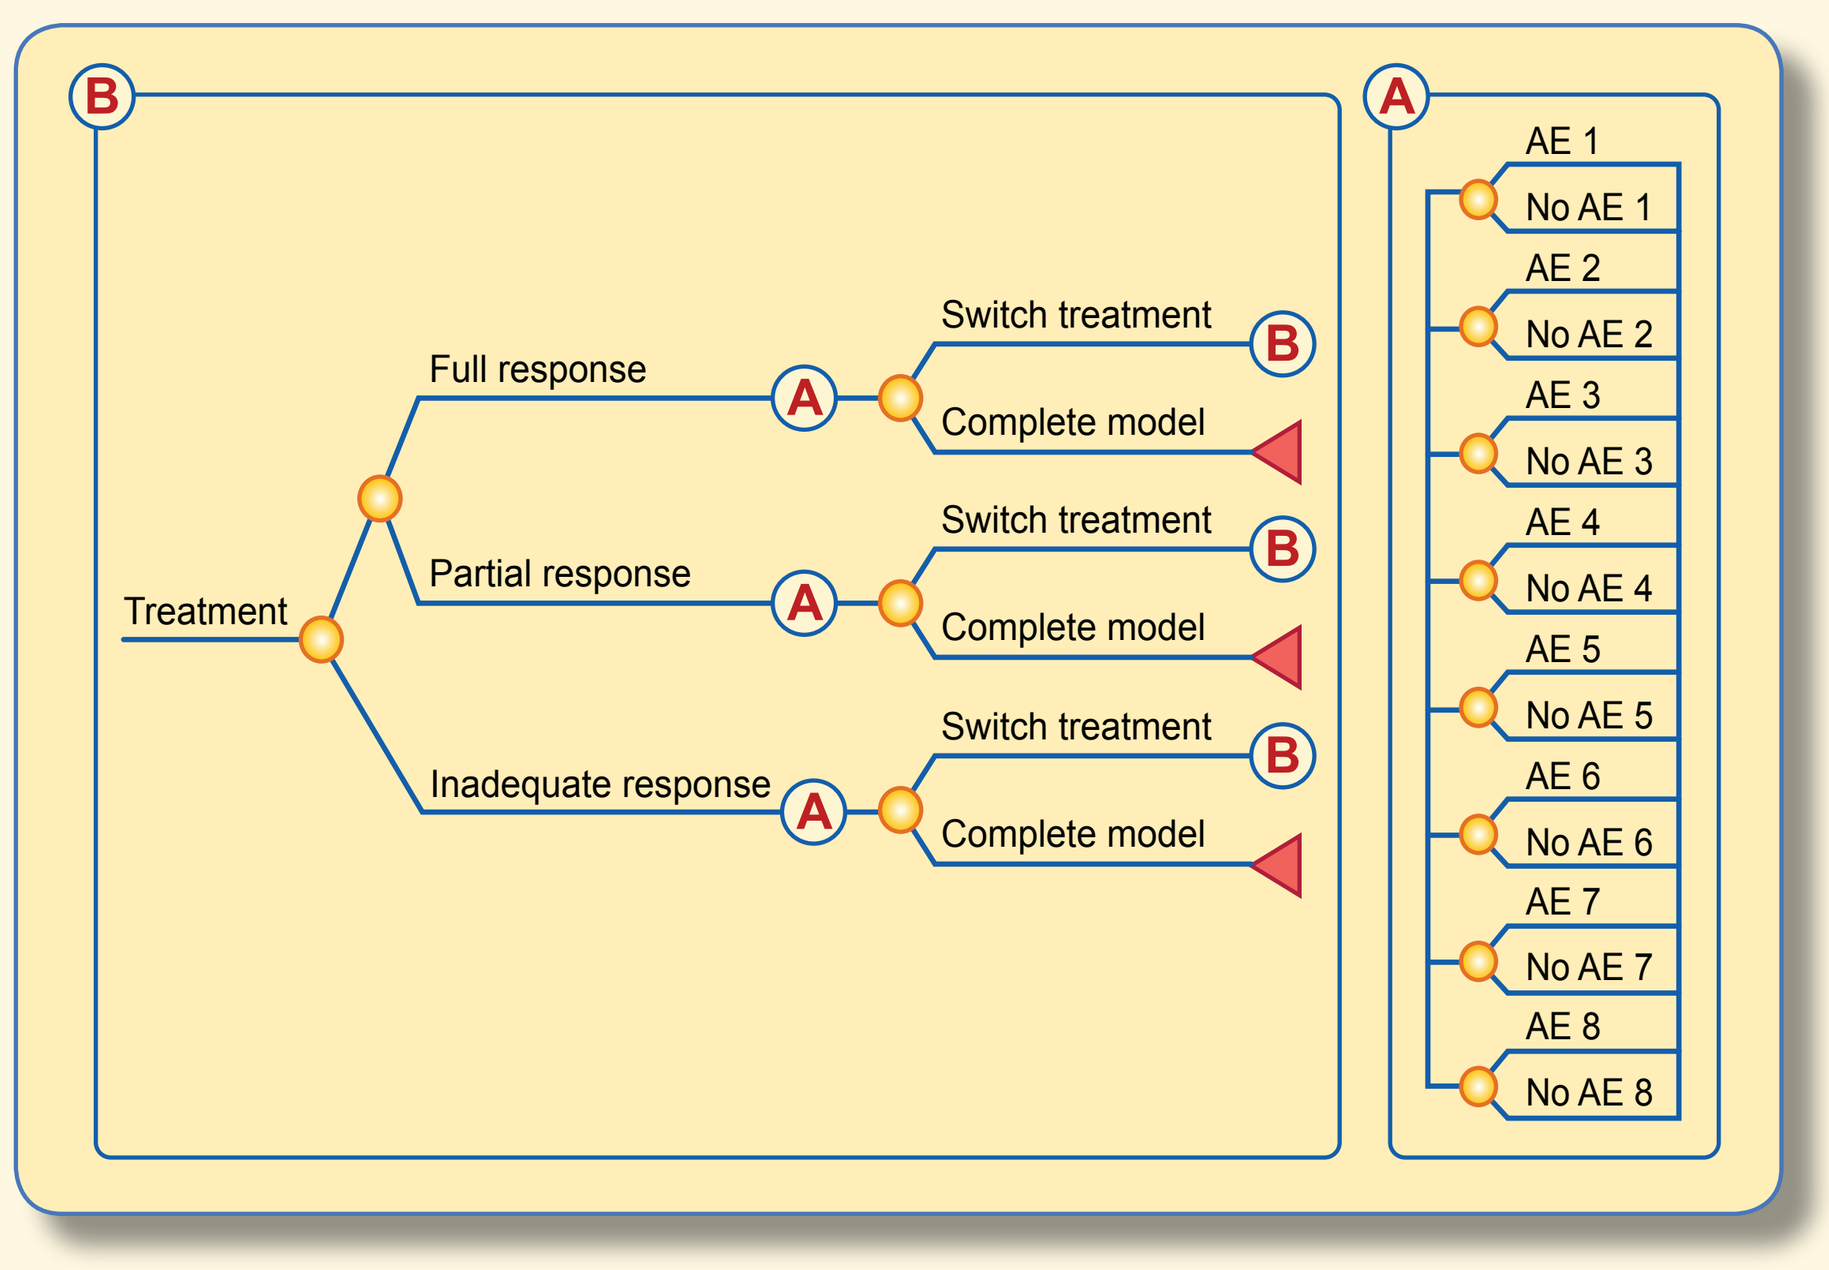

B
Treatment
Full response
Partial response
Inadequate response
A
Switch treatment
Complete model
B
A
AE 1
No AE 1
AE 2
No AE 2
AE 3
No AE 3
AE 4
No AE 4
AE 5
No AE 5
AE 6
No AE 6
AE 7
No AE 7
AE 8
No AE 8

- Figure 3 depicts the potential comparators to the new drug, at each potential line of therapy.

Figure 3. Potential Comparators, by Indication and Line of Therapy

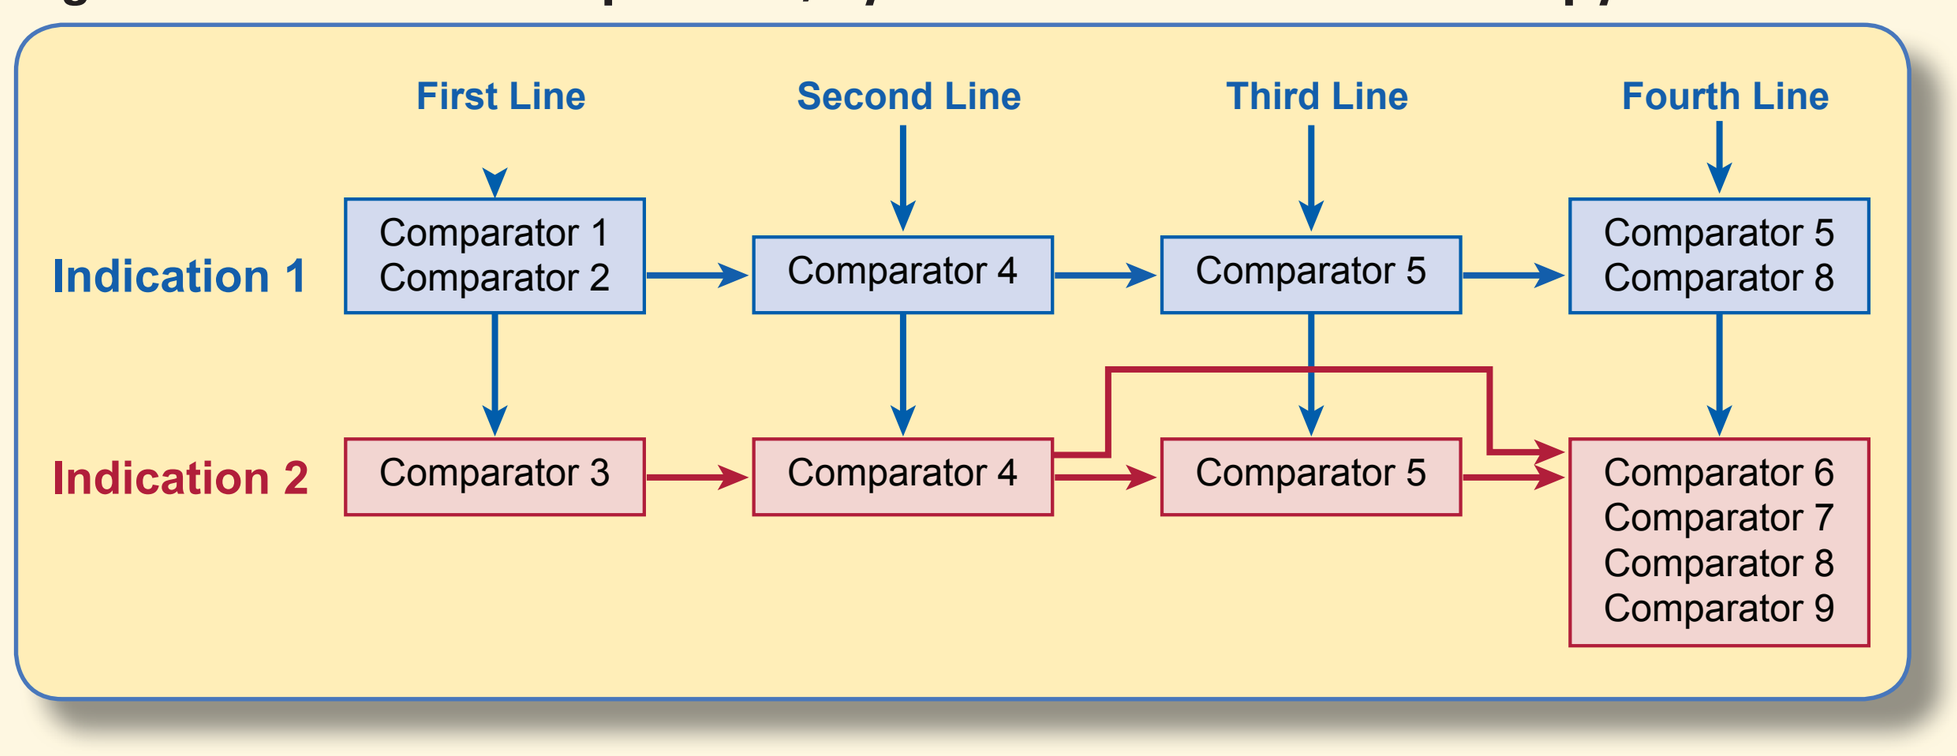

First Line
Second Line
Third Line
Fourth Line
Indication 1
Comparator 1
Comparator 2
Comparator 4
Comparator 5
Comparator 5
Comparator 8
Indication 2
Comparator 3
Comparator 4
Comparator 5
Comparator 6
Comparator 7
Comparator 8
Comparator 9

- Using the threshold value-based pricing model, it was possible to estimate the value-based price opportunities available for the drug. Table 3 presents these hypothetical opportunities for the drug.

Table 3. Value-Based Price Opportunities for New Drug (Price per Day), by Line of Therapy, Comparator, and Whether Target or Ideal Product Profile Was Evaluated for Each Indication

Line of Therapy	Comparator	Indication 1		Indication 2	
		Target	Ideal	Target	Ideal
First line	Comparator 1	£8.75	£10.61	NA	NA
	Comparator 2	£10.00	£12.00	NA	NA
	Comparator 3	NA	NA	£11.50	£12.47
Second line	Comparator 4	£12.00	£13.00	£9.02	£9.85
Third line	Comparator 5	£24.31	£26.01	£23.92	£24.00
Fourth line	Comparator 5	£16.05	£17.09	NA	NA
	Comparator 6	NA	NA	£9.79	£10.56
	Comparator 7	NA	NA	£6.81	£7.62
	Comparator 8 (Drug B)	£10.50	£10.96	£11.28	£11.50
	Comparator 9	NA	NA	£12.49	£13.31

NA = not applicable.

- Comparator 8 was the currently marketed drug to which the new drug was expected to be similar, and served as the basis of the draft product profile benchmarks.
- However, the threshold value-based pricing exercise suggested that even if the new drug achieved the ideal product profile targets, it would not be able to achieve the target price (a 20% premium over Comparator 8, priced at £10 per day).
- The threshold value-based pricing exercise suggested that the new drug may have a better value-based price opportunity (reflecting greater unmet need and/or ability of the new drug to provide better health benefit) if it were developed instead as a third-line option in comparison with Comparator 5.
- It was possible to use the threshold pricing model to examine the influence of each product attribute on the estimated value-based price opportunity in any given indication-line of therapy-comparator scenario.
- Figure 4 depicts the influence of AE 1 on maximum value-based price in indication 1, third-line, versus comparator 5 (the scenario above that indicates the best overall potential value of the new drug).
- The difference between the target price and the maximum value-based price opportunity at the base case AE 1 incidence rate indicates the potential missed value-based price opportunity.

Figure 4. Influence of AE 1 on Value-Based Price of New Drug

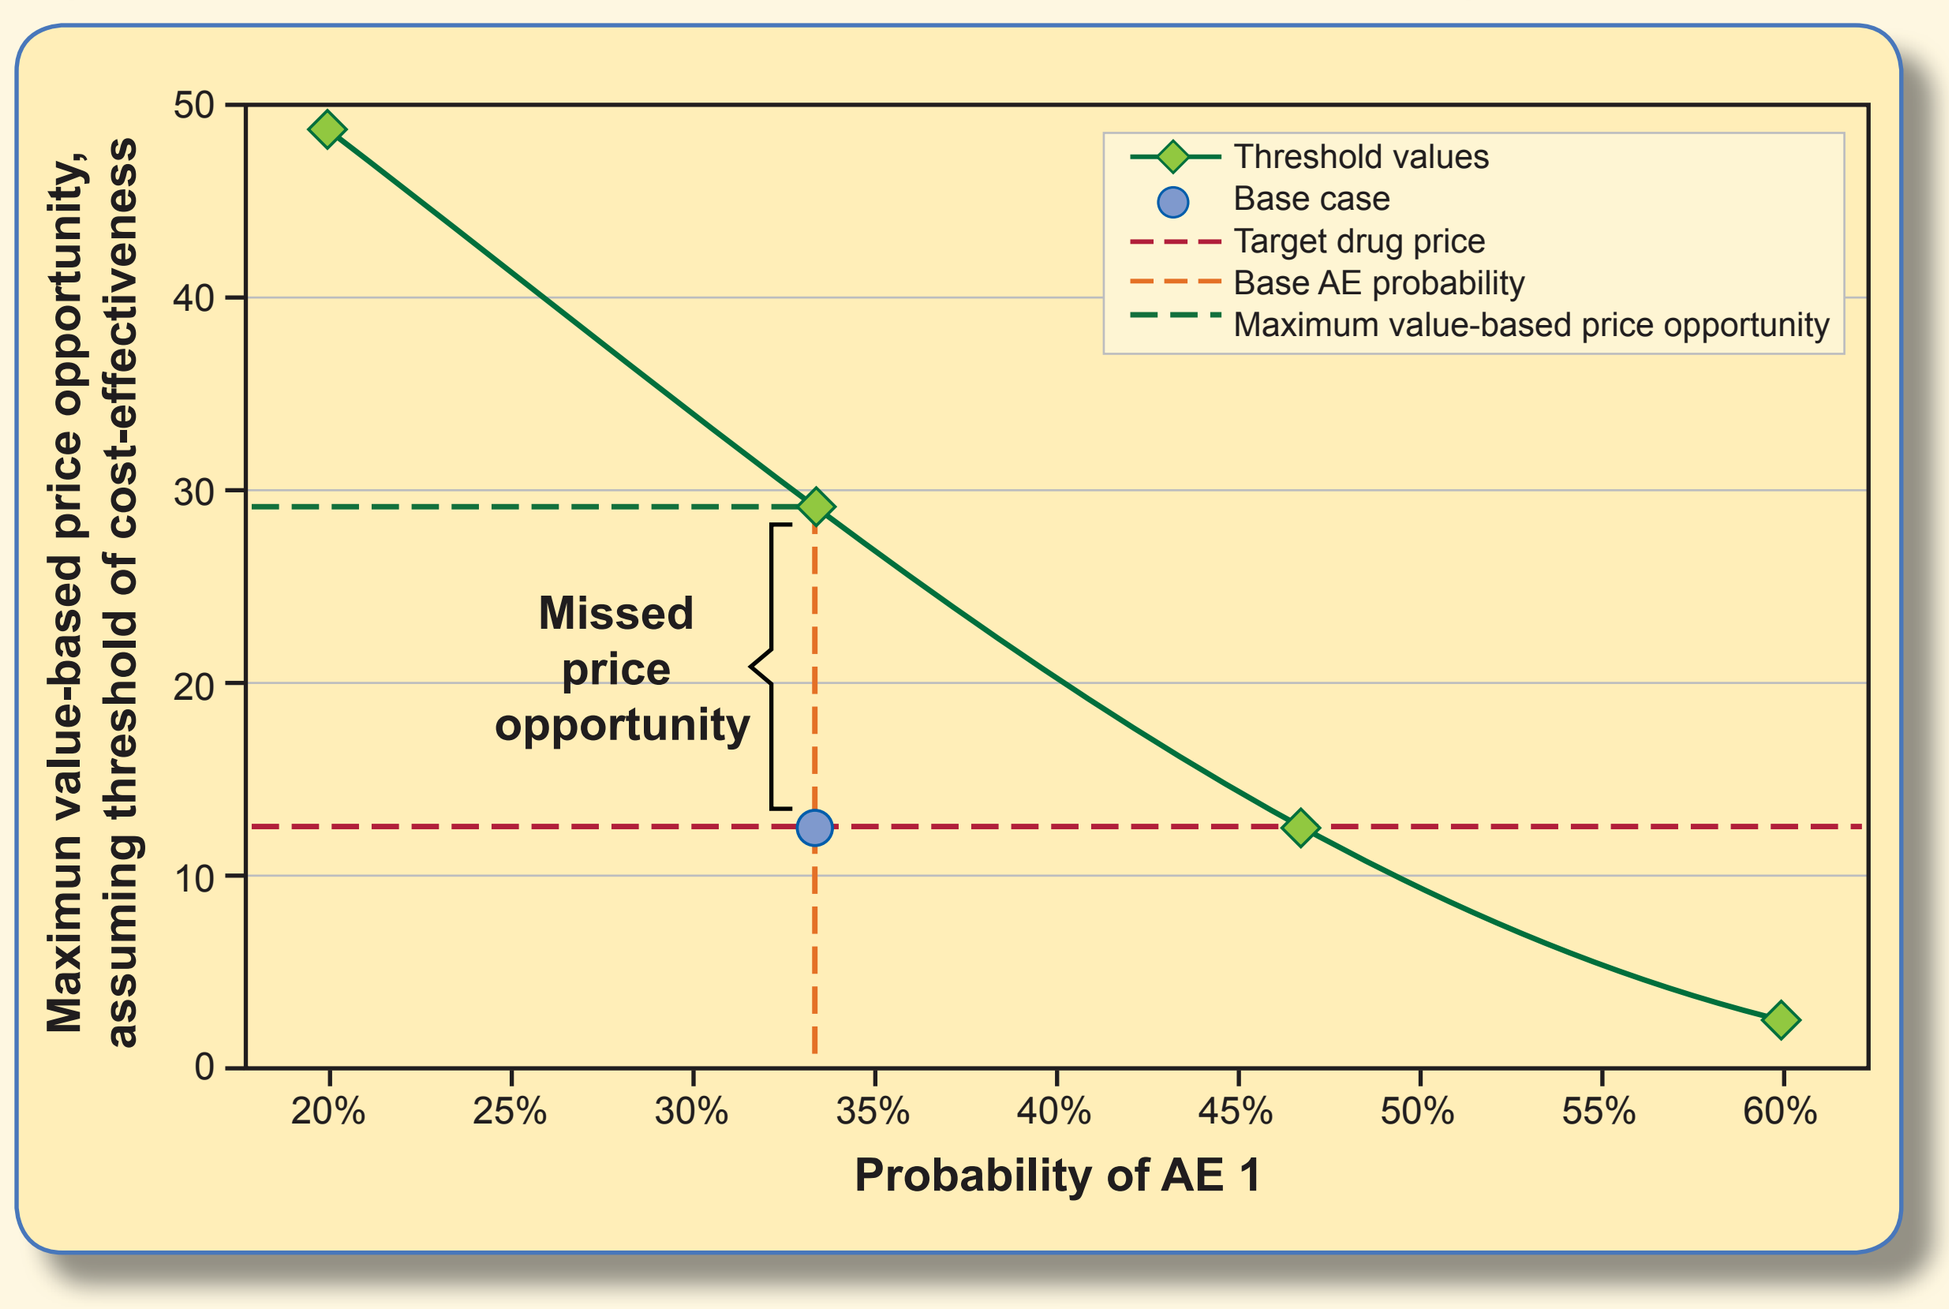

Maximun value-based price opportunity, assuming threshold of cost-effectiveness
50
40
30
20
10
0
20%
25%
30%
35%
40%
45%
50%
55%
60%
Probability of AE 1
Threshold values
Base case
Target drug price
Base AE probability
Maximum value-based price opportunity
Missed price opportunity

Figure 5. Scenario in Which New Drug is Cost-Saving

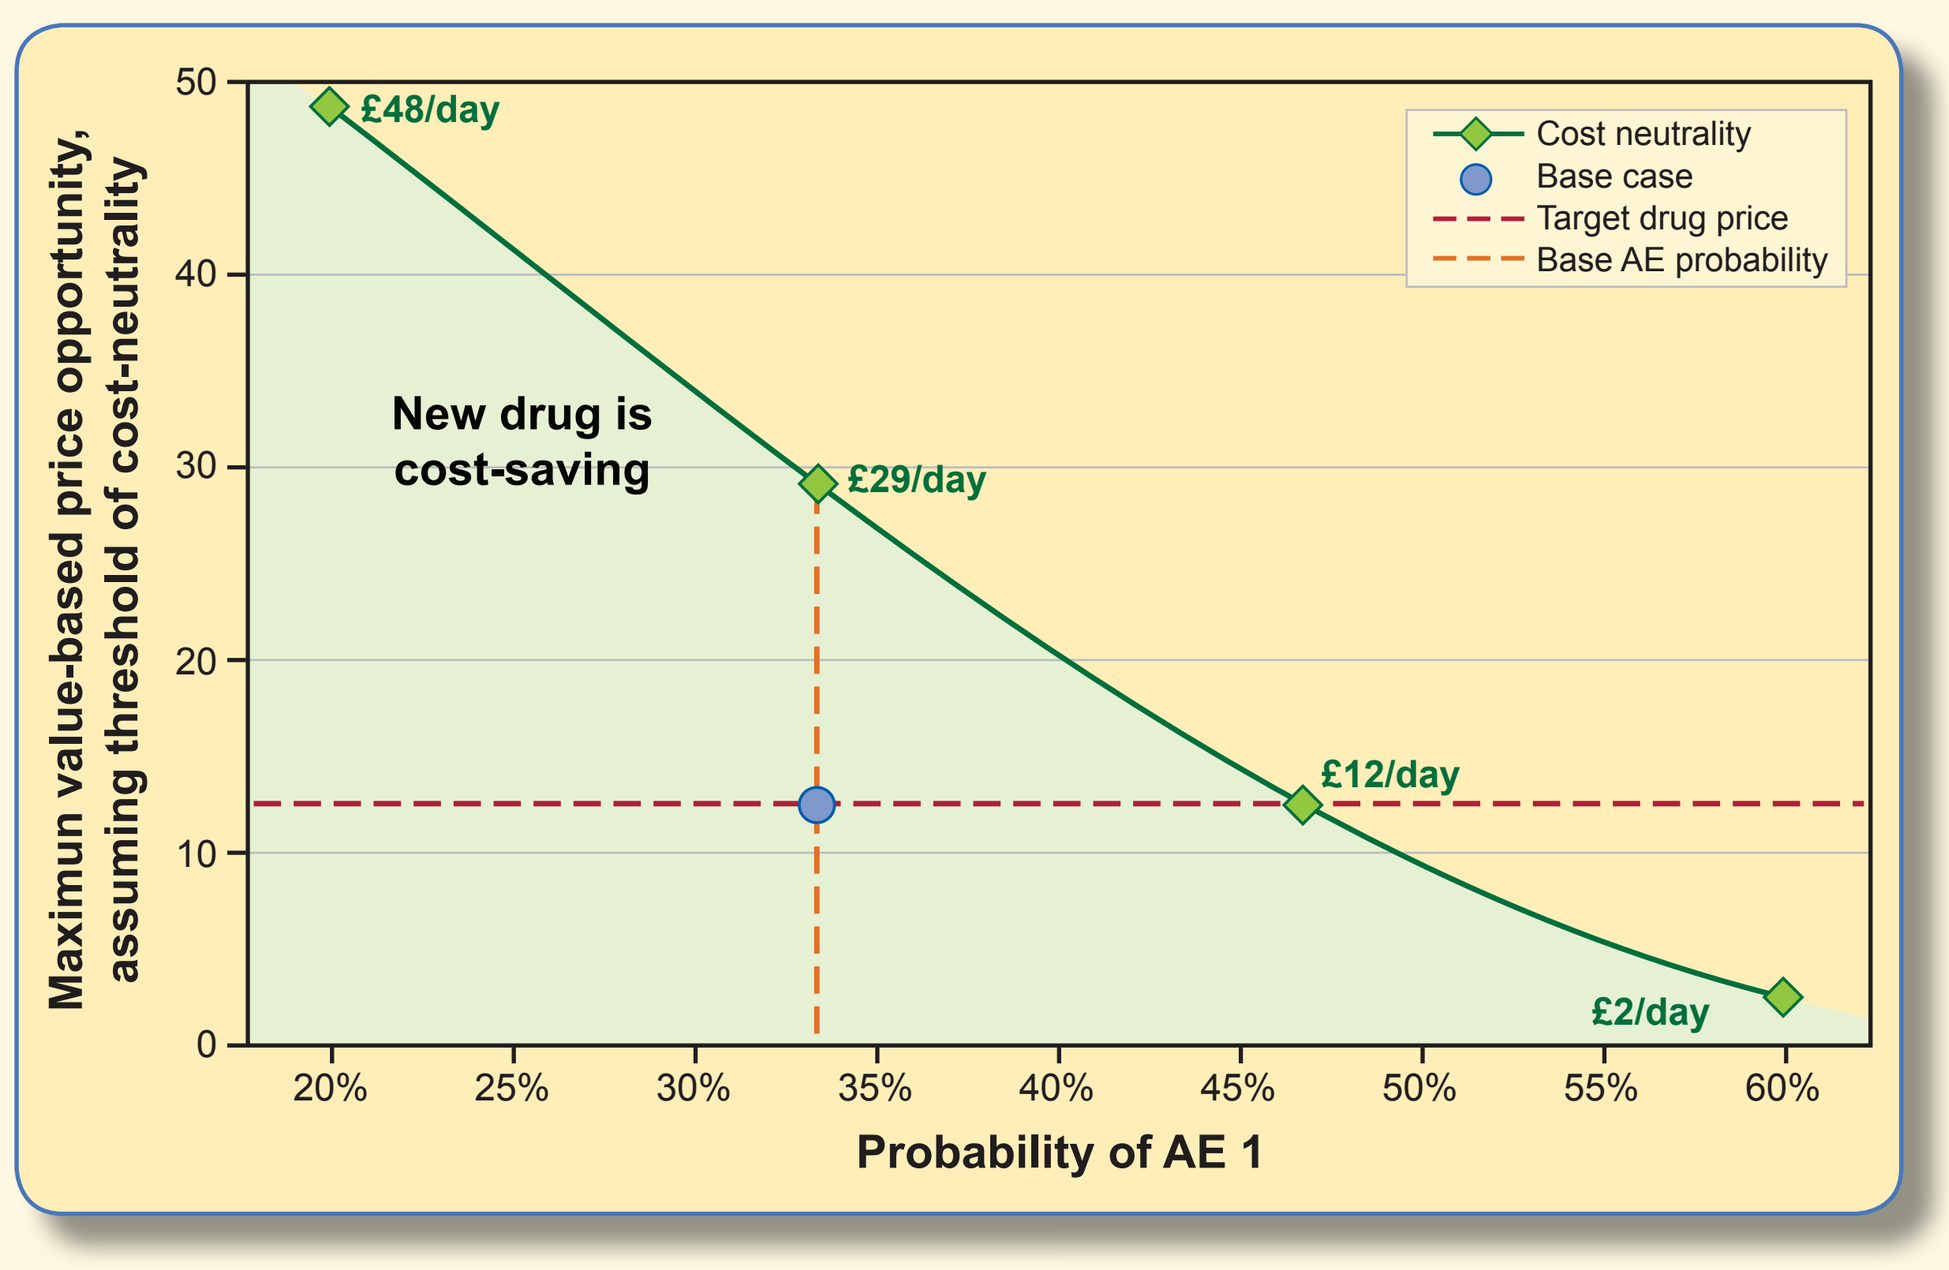

Maximun value-based price opportunity, assuming threshold of cost-neutrality
50
40
30
20
10
0
20%
25%
30%
35%
40%
45%
50%
55%
60%
Probability of AE 1
£48/day
£29/day
£12/day
£2/day
New drug is cost-saving
Cost neutrality
Base case
Target drug price
Base AE probability

- It is possible to do one-way, multiway and probabilistic sensitivity analyses on estimates of value-based price opportunities.[1]

CONCLUSIONS

- A threshold pricing model is a powerful early decision tool for helping drug manufacturers construct a value-driven development plan and a value-based price strategy.
- A threshold pricing model also may be useful for bringing clarity to a new product development team; quantifying, testing, and clarifying expectations of new product performance; and achieving target price and adequate market access.
- Constructed appropriately, threshold pricing models can be used to prioritize among possible indications, identify target subpopulations, select the appropriate line of therapy, and choose and clarify required performance against comparators.

REFERENCE

1. Mladsi D, Earnshaw S, Akashi N, Keith M. Proposed methods for conducting sensitivity analyses on threshold-derived estimates of value-based price and product profiles of early stage drugs. Poster presented at the ISPOR 13th Annual European Conference; November 6-9, 2010. Prague, Czech Republic.

DISCLOSURE

Financial support was provided by Shire Pharmaceuticals.

CONTACT INFORMATION

Deirdre Mladsi
Head, Health Economics and Market Access

RTI Health Solutions
200 Park Offices Drive
Research Triangle Park, NC 27709-2194

Phone: +1.919.541.7094
Fax: +1.919.541.7222
E-mail: dmladsi@rti.org

Presented at: ISPOR 13th Annual European Conference
November 6-9, 2010
Prague, Czech Republic