RTI(h)(s)

Cost-Effectiveness of Rasagiline Versus Ropinirole Extended Release in Delaying Levodopa in the Treatment of Early Parkinson's Disease in the United States

Raymond A Farkouh,[1] Michele R Wilson,[1] Marcy L Tarrants,[2] Jane Castelli-Haley,[2] Christophe Armand[3]

[1] RTI Health Solutions, Research Triangle Park, NC, United States

[2] Teva Neuroscience, Kansas City, MO, United States

[3] H. Lundbeck A/S, Paris, France

BACKGROUND

- Parkinson's disease (PD) is a common disease that affects approximately 1 million people in the United States (US) (Olanow and Koller, 1998).
- The incidence of PD is approximately 60,000 new cases per year (Olanow and Koller, 1998).
- The disease is associated with limitations in physical function and autonomy, and leads to severe disability.
- As PD progresses, patients and their families experience substantial health and economic burdens.
- Pharmacologic intervention is available for PD. Possible treatment options include:
 - Levodopa (LD): gold standard for controlling motor symptoms of PD.
 - Avoided as first-line treatment due to the long-term irreversible motor complications (dyskinesia) it induces (Jankovic, 2005). Dyskinesia is involuntary movement interfering with normal functioning.
 - Concerns about LD efficacy decay and dyskinesia have given rise to a growing consensus not to start LD as first-line treatment in patients younger than 70 years.
 - Dopamine agonist (DA): available as first-line therapy; the most common pharmacologic strategy other than LD.
 - Administration is more costly than LD treatment (Weiner, 2004) and exposes patients to serious adverse events such as psychiatric disorders, cardiovascular fibrosis, and sleep attacks.
 - Expected to induce fewer motor fluctuations than LD (Rascol et al., 2000); however, DA-induced dyskinesias do occur. We conservatively assume that no DA-induced dyskinesias occur.
 - Ropinirole XL (Requip XL, GlaxoSmithKline) is a new once-daily formulation.
 - Rasagiline mesylate (rasagiline [Azilect, Teva Neurosciences]): available as first-line therapy; a once-daily, selective irreversible monoamine oxidase type-B inhibitor.
 - Administration is more costly than LD treatment; however, has a favorable tolerability profile.
 - Not associated with motor fluctuations in monotherapy use.
- Because LD-induced dyskinesias are linked to poor quality of life and higher health care costs (Péchevis et al., 2005), postponing the appearance of disabling motor complication could be an effective strategy for reducing costs associated with PD.

OBJECTIVE

The purpose of this study was to evaluate the cost-effectiveness of initiating first-line treatment of early PD with once-daily rasagiline monotherapy compared with initiating therapy with a once-daily DA, specifically ropinirole XL, in delaying appearance of LD-induced dyskinesias in US patients with early-stage PD.

METHODS

Model Structure

- To model the delay of initiating LD therapy, a Markov model (Figure 1) was developed to evaluate the costs and outcomes of the two early PD treatment strategies.

Figure 1. Markov Model: Early PD Treatment Pathways[a]

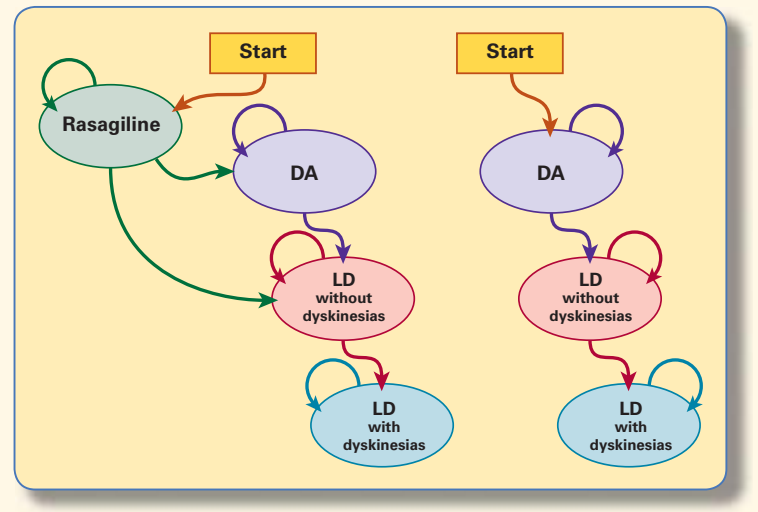

Start
Start
Rasagiline
DA
DA
LD without dyskinesias
LD without dyskinesias
LD with dyskinesias
LD with dyskinesias

[a] All states may transition to death.

- Patients can transition therapy every 6 months.
- The model time horizon is 5 years (consistent with the long-term follow-up from the TVP-1012 in Early Monotherapy for Parkinson's Disease Outpatients [TEMPO] trial [Hauser et al., 2009] and the pivotal ropinirole trial [Rascol et al., 2000]).
- Patients taking rasagiline could switch to a DA because rasagiline has a mode of action distinct from DAs and a decrease in rasagiline efficacy should not prevent switching to a DA.
- The model does not include combining therapies; instead, switching therapies was determined to be the most conservative modeling strategy.
- The primary outcomes of interest over a 5-year time horizon in US patients with early-stage PD are:
 - Time to LD
 - Time to LD-induced dyskinesias
 - Total costs
 - Total quality-adjusted life-years (QALYs)
 - Incremental cost per QALY
- The model is presented from a US managed care perspective.
- Costs and outcomes were discounted at 3% per annum.

Patient Characteristics

We examined a patient diagnosed with early PD who did not require LD for their condition. The patient starts in Hoehn and Yahr (H&Y) stage 1.5 with an average age of 61 years, as was observed in Hauser et al. (2009).

TEMPO was a 6-month double-blind, placebo-controlled study of 404 patients with early PD randomized to rasagiline 1 or 2 mg/day or placebo, followed by a blind active extension, where patients under placebo were switched to rasagiline 1 mg/day.

Transition Probabilities

Table 1 summarizes health state transition probabilities.

Table 1. Time-Specific Transition Probabilities

Cycle	Rasagiline to DA	Rasagiline to LD Without Dyskinesias	DA to LD Without Dyskinesias	LD to LD With Dyskinesias	Any State to Death
1	0.0400	0.0300	0.1020	0.1000	0.0243
2	0.1600	0.0900	0.1020	0.0444	0.0255
3	0.2200	0.0900	0.1020	0.0465	0.0267
4	0.1400	0.0300	0.1020	0.0600	0.0279
5	0.1200	0.0700	0.1020	0.1178	0.0291
6	0.1600	0.0500	0.1020	0.0600	0.0303
7	0.2000	0.0500	0.1020	0.0600	0.0315
8	0.0600	0.0000	0.1020	0.1678	0.0327
9	0.0400	0.1000	0.1020	0.1000	0.0340
10	0.0800	0.0800	0.1020	0.0889	0.0353
Source	Hauser et al., 2009	Hauser et al., 2009	Rascol et al., 2000	Rascol et al., 2000	Kung et al., 2008; Clarke, 1995

Details for time-specific transition probabilities:

- Rasagiline to DA
 - Transition probabilities from rasagiline to ropinirole XL were calculated from Hauser et al. (2009). Time-dependent transition probabilities were calculated from the percentage of patients starting any DA (selected by the physician) during each 6-month cycle following the start of rasagiline therapy.
 - The DA was assumed to be ropinirole XL because the effective clinical outcome was the requirement for a drug pertaining to that pharmacologic class.
- Rasagiline to LD without dyskinesias
 - Transition probabilities from rasagiline to LD were calculated from Hauser et al. (2009). Time-dependent transition probabilities were calculated from the percentage of patients starting LD during each 6-month cycle following the start of rasagiline therapy.
- DA to LD without dyskinesias
 - The transition probabilities from DA to LD were calculated from Rascol et al. (2000).
 - The 6-month transition probability was calculated from the percentage of patients receiving supplemental LD within 5 years of treatment with ropinirole, where 65.88% (56 out of 85) of patients received supplemental LD by the end of the 5-year period due to such factors as adverse effects of DA therapy or poor response to DA monotherapy.
 - Probability of transition from DA to LD in 6 months is:

$$\left(1-e^{\left(\frac{\ln\left(1-\frac{56}{85}\right)}{\frac{5}{0.5}}\right)}\right)=0.1020$$

- LD without dyskinesias to LD with dyskinesias
 - The probability of developing dyskinesias while on LD therapy was obtained from Figure 2 of Rascol and colleagues' (2000) 5-year study of the incidence of dyskinesia in patients with early PD, focusing on the patients treated with LD.
 - The Kaplan-Meier survival curve was converted to time-dependent transition probabilities to reflect the remaining sample of patients free of dyskinesias:

$$\text{Transition probability}_t = 1 - [\text{cumulative survival}_{t-1} / \text{cumulative survival}_t]$$

- All health states allow a transition to death
 - The transition probabilities are based on US population-level gender- and age-specific all-cause mortality (Kung et al., 2008) assuming a starting age of 61 years.
 - A PD-specific relative mortality adjustment of 2.3 (Clarke, 1995) is applied to the all-cause mortality to derive a PD-specific mortality probability.
 - The probability of death increases each cycle to reflect the aging population over the 5-year course of the model.
 - Risk of death is assumed independent of current treatment strategy.

Costs

Table 2 presents specific pharmaceutical costs, nonpharmaceutical direct medical costs, and utility weights.

Table 2. Model Inputs for Health States

Health State	Pharmaceutical Costs[a]	Nonpharmaceutical Direct Medical Costs[a]	Utility Weights
Rasagiline	$1,506.00	$8,520.53	0.83
DA	$1,171.50 (cycle 1) $1,757.25 (cycle 2) $2,343.00 (cycles 3-10)	$8,520.53	0.83
LD without dyskinesias	$497.37	$8,520.53	0.72
LD with dyskinesias	$497.37	$14,304.01	0.48

[a] Per cycle.

Health State Costs

- Orsini et al. (2004) performed a database analysis of Medstat's Marketscan Commercial Claims and Encounters or Medicare Coordination of Benefits claims database. The mean age of the sample was 73 years with 44% women.
- Outpatient pharmaceutical costs were removed from the total per patient costs. The remaining cost was inflated to 2007 US dollars (US Department of Labor: Bureau of Labor Statistics, 2008) and divided by 2 to yield the per patient per 6 month cycle nonpharmaceutical direct medical costs.
- These were assumed to be the same across all nondyskinetic health states.

Dyskinesia Cost Multiplier

Patients with dyskinesias exhibited higher direct medical costs (Péchevis et al., 2005). The effect of dyskinesias on health-related direct costs was obtained from a 6-month observational study (Péchevis et al., 2005) conducted in three European countries of patients at various stages of PD. Only costs incurred by patients with a UPDRS (Unified Parkinson's Disease Rating Scale) IVa (dyskinesia) score comprised between 1 and 8 were considered. Total nonpharmaceutical direct medical costs were multiplied by 1.679 to obtain a relative cost for those with LD-induced dyskinesias.

Pharmaceutical Costs

- Rasagiline
 - Rasagiline was assumed to be dosed at 1 mg once daily.
 - $229.22 (wholesale acquisition cost [WAC]) for 30 pills, national drug code (NDC): 68546-0229-56 (Red Book, 2008).
- DA
 - Ropinirole XL was assumed to be dosed according to an escalating dosing schedule where patients consume 8 mg/day in the first 6 months, 12 mg/day in the subsequent 6 months, and 16 mg/day after the first year.
 - $195.25 (WAC) for 30 pills of 8 mg per pill, NDC: 00007-4888-13 (Red Book, 2008). This calculates to $0.8135 per mg.
- LD
 - LD was assumed to be a coformulation of carbidopa (CD) and LD, using a 1:4 ratio of CD to LD (Hoerger et al., 1998) and an LD dosage of 400 mg/day (Hoerger et al., 1998). As CD and LD are generic, we pulled 45 Red Book entries that satisfied these criteria, and calculated an average cost per cycle according to the following:

$$\frac{\text{180 days per cycle} \times \text{Cost per package (AWP)} \times \text{\# pills per day to obtain 400 mg LD}}{\text{\# pills per package}}$$

Utility

- QALYs were calculated by associating a utility weight to each health state, which is represented by the current treatment.
- Utility weights were obtained from Palmer et al. (2000). Palmer presented both visual analog scale (VAS) and standard gamble (SG) approaches to deriving health-state values. We used the VAS values in the base case. Table 2 presents the utility weights used in each modeled health state.
 - The rasagiline and DA health states assumed patients were at an H&Y stage 1.5, with no off time.
 - The LD without dyskinesias health state assumed patients were at an H&Y stage 2.5, with no off time.
 - The LD with dyskinesias health state assumed patients were at an H&Y stage 2.5, with off time, and a weighted average utility was calculated for those patients in the health state, assuming dyskinesias were correlated with Palmer's classification of off-time motor fluctuations.

RESULTS

Base Case Results

Results (Table 3) showed that initiating treatment with rasagiline is dominant (lower costs and higher QALYs) to initiating treatment with ropinirole XL.

Table 3. Results Over 5 Years of Early PD Treatment by First-Line Therapy

Outcomes	Rasagiline	Ropinirole XL	Incremental
Total cost (US $)	$83,599.26	$85,259.50	-$1,660.24
QALYs	3.2101	3.1493	0.0608
Time to LD (years)	2.9663	2.5952	0.3711
Time to dyskinesia (years)	3.8770	3.7937	0.0833
Time on LD free of dyskinesias (years)	0.9107	1.1985	-0.2878
Incremental cost per QALY	Initiating treatment with rasagiline is dominant (lower costs and higher QALYs)		

Sensitivity Analysis Results

Figure 2 displays one-way sensitivity analysis results, which show that initiating treatment with rasagiline remained cost-savings in nearly all sensitivity analyses.

Figure 2. One-Way Sensitivity Analysis Results

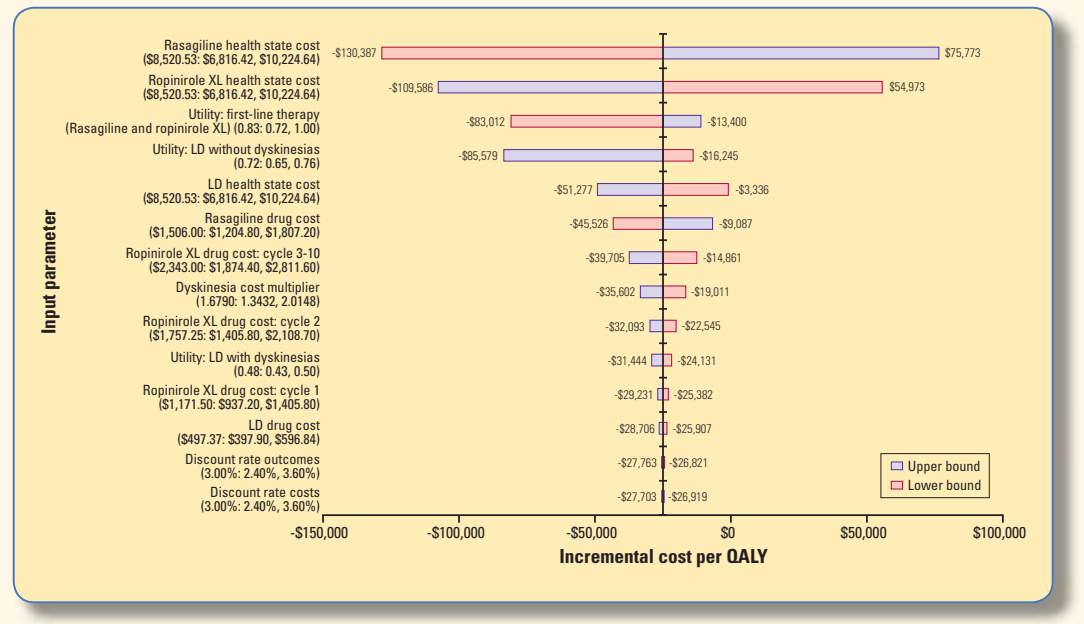

Rasagiline health state cost ($8,520.53: $6,816.42, $10,224.64)
-$130,387
$75,773
Ropinirole XL health state cost ($8,520.53: $6,816.42, $10,224.64)
-$109,586
$54,973
Utility: first-line therapy (Rasagiline and ropinirole XL) (0.83: 0.72, 1.00)
-$83,012
-$13,400
Utility: LD without dyskinesias (0.72: 0.65, 0.76)
-$85,579
-$16,245
LD health state cost ($8,520.53: $6,816.42, $10,224.64)
-$51,277
-$3,336
Rasagiline drug cost ($1,506.00: $1,204.80, $1,807.20)
-$45,526
-$9,087
Ropinirole XL drug cost: cycle 3-10 ($2,343.00: $1,874.40, $2,811.60)
-$39,705
-$14,861
Dyskinesia cost multiplier (1.6790: 1.3432, 2.0148)
-$35,602
-$19,011
Ropinirole XL drug cost: cycle 2 ($1,757.25: $1,405.80, $2,108.70)
-$32,093
-$22,545
Utility: LD with dyskinesias (0.48: 0.43, 0.50)
-$31,444
-$24,131
Ropinirole XL drug cost: cycle 1 ($1,171.50: $937.20, $1,405.80)
-$29,231
-$25,382
LD drug cost ($497.37: $397.90, $596.84)
-$28,706
-$25,907
Discount rate outcomes (3.00%: 2.40%, 3.60%)
-$27,763
-$26,821
Discount rate costs (3.00%: 2.40%, 3.60%)
-$27,703
-$26,919
Input parameter
Upper bound
Lower bound
-$150,000
-$100,000
-$50,000
$0
$50,000
$100,000
Incremental cost per QALY

Table 4 presents scenario analyses.

Table 4. Scenario Analyses

Analysis Name	Result
SG utility values	Rasagiline strategy dominates
Transition probability: lower bound of DA to LD[a]	Rasagiline strategy dominates
Transition probability: upper bound of DA to LD[b]	Rasagiline strategy dominates
Transition probability: lower bound of LD without dyskinesias to LD with dyskinesias[c]	Rasagiline strategy dominates
Transition probability: upper bound of LD without dyskinesias to LD with dyskinesias[c]	Rasagiline strategy dominates
Transition probability: lower bound of rasagiline to DA[c]	Rasagiline strategy dominates
Transition probability: upper bound of rasagiline to DA[c]	Rasagiline strategy dominates
Transition probability: lower bound of rasagiline to LD[c]	Rasagiline strategy dominates
Transition probability: upper bound of rasagiline to LD[c]	Rasagiline strategy dominates

[a] Lower bound calculated from the intent to treat population, where 92 out of 179 patients received LD (Rascol et al., 2000).

[b] Upper bound calculated by assuming that all patients who withdrew prematurely from the clinical trial required LD: 94 withdrew and 56 received LD out of 179 patients over 5 years (Rascol et al., 2000).

[c] Lower bound and upper bound assumes a 50% relative decrease and increase respectively in the time-dependant transition probability.

CONCLUSIONS

Although this study compares once-daily PD treatment options, additional cost-effectiveness analyses of all US Food and Drug Administration–approved early PD treatment comparators is necessary and currently underway to further define the most cost-effective treatment paradigm for early PD.

For once-daily PD treatment options, this model indicates that initiating early PD treatment with rasagiline delayed treatment with LD and subsequent LD-induced dyskinesias, saved costs, and resulted in more QALYs. Rasagiline is therefore a dominant strategy when compared with initiating early PD treatment with ropinirole XL.

REFERENCES

Azilect Prescribing Information. 2006. Available at: http://www.azilect.com/PrescribingInformation.pdf.ashx. Accessed March 24, 2009.

Clarke CE. Does levodopa therapy delay death in Parkinson's disease? A review of the evidence. Mov Disord 1995;10:250-6.

Hauser RA, Lew MF, Hurtig HI, Ondo WG, Wojcieszek J, Fitzer-Attas CJ; on behalf of the TEMPO Open-label Study Group. Long-term outcome of early versus delayed rasagiline treatment in early Parkinson's disease. Mov Disord 2009;24(4):564-73.

Hoerger TJ, Bala MV, Rowland C, Greer M, Chrischilles EA, Holloway RG. Cost effectiveness of pramipexole in Parkinson's disease in the US. Pharmacoeconomics 1998;14:541-57.

Jankovic J. Motor fluctuations and dyskinesias in Parkinson's disease: clinical manifestations. Mov Disord 2005;20(Suppl 11):S11-6.

Kung H, Hoyert DL, Xu J, Murphy SL. Deaths: final data for 2005. National Vital Statistics Reports 56(10). Hyattsville, MD: National Center for Vital Statistics; 2008.

Olanow CW, Koller WC. An algorithm (decision tree) for the management of Parkinson's disease: treatment guidelines. Neurology 1998;50(Suppl 3):S1–S57.

Orsini L, Castelli-Haley J, Kennedy S, Huse D. Healthcare utilization and expenditures among privately insured patients with Parkinson's disease in the U.S. Poster presented at the 8th Annual Meeting of the Movement Disorder Society's International Congress, Rome, Italy. Jun 2004.

Palmer CS, Schimer JK, Snyder E, Scott B. Patient preferences and utilities for 'off-time' outcomes in the treatment of Parkinson's disease. Qual Life Res 2000;9:819-27.

Péchevis M, Clarke CE, Vieregge P, Khoshnooda B, Deschaseaux-Voineta B, Berdeaux G, Ziegler M. Effects of dyskinesias in Parkinson's disease on quality of life and health related costs: a prospective European study. Eur J Neurol 2005;12:956–63.

Rascol O, Brooks DJ, Korczyn AD, et al. A five-year study of the incidence of dyskinesia in patients with early Parkinson's disease who were treated with ropinirole or levodopa. 056 Study Group. N Engl J Med 2000;342:1484-91.

Red Book for Windows, Version 61127, Vol. 50. Thomson PDR, Montvale, NJ. Oct 2008.

Requip XL Prescribing Information. 2008. Available at: http://us.gsk.com/products/assets/us_requipxl.pdf. Accessed March 24, 2009.

US Department of Labor: Bureau of Labor Statistics. Consumer price index – all urban consumers (current series). 2008. Available at: http://data.bls.gov/PDQ/outside.jsp?survey=cu. Accessed April 1, 2009.

Weiner WJ. Initial treatment of Parkinson disease: levodopa or dopamine agonists. Arch Neurol 2004;61:1966-9.

CONTACT INFORMATION

Raymond Farkouh
Associate Director, Health Economics

RTI Health Solutions
200 Park Offices Drive
Research Triangle Park, NC 27709 United States

Telephone: 919-541-7332
Fax: 919-541-7222
E-mail: rfarkouh@rti.org

Presented at: ISPOR 14th Annual International Meeting
May 16-20, 2009
Orlando, Florida, United States