Assessing the Impact of Smoking Cessation Therapies on a Managed Care Organization's Budget using Census Region and State-Specific Smoking Estimates

Graham CN,[1] Earnshaw SR,[1] Saunders M[2]

[1]RTI Health Solutions, Research Triangle Park, North Carolina, United States

[2]GlaxoSmithKline Consumer Healthcare, Moon Township, Pennsylvania, United States

BACKGROUND

- Smoking is one of the mostly costly addictions in the United States (US) today.[1]
- The prevalence of smoking in the US varies greatly by state and by census region analyzed.[2]
- Stopping smoking can translate to long-term cost savings for former smokers and managed care organizations (MCOs).[3, 4, 5, 6, 7]
- Quitting smoking is difficult—only 3% to 5% of quit attempts are successful over the long term (e.g., 6-12 months).[8, 9]
- Because of the difficulty in quitting, introducing new pharmacological therapies is valuable. However, history has shown that when a new smoking cessation therapy is introduced to the market, there is a dramatic uptake of the new therapy.[10]
- As new smoking cessation therapies come onto the market, MCOs need to be prepared with accurate financial planning in order to appropriately estimate the impact on their prescription drug budget.

OBJECTIVES AND PURPOSE

In this study, we estimate the potential budget impact of adding a new smoking cessation therapy to an existing mix of covered and noncovered therapies in an MCO's formulary. This analysis is undertaken at both the state level and census region level to provide an accurate representation of the budget impact for MCOs across differing geographic areas. This analysis can inform decision makers who are faced with the prospect of an increasing number of smoking cessation therapies and are considering adding new therapies to their formularies.

CONTACT INFORMATION

Christopher N. Graham, MSc

RTI Health Solutions, RTI International
3040 Cornwallis Road, PO Box 12194
Research Triangle Park, NC 27709-2194

Phone: 919.541.6322
E-mail: cgraham@rti.org

Presented at:
The Academy of Managed Care Pharmacy (AMCP)
19th Annual Meeting Showcase, San Diego, CA
April 11-14, 2007

METHODS

To evaluate the potential economic impact by state and census region of the use of new and existing smoking cessation therapies on an MCO's budget, we modeled 1-year budgetary outcomes in a decision-analytic framework (Figure 1).

- Two scenarios were modeled: (1) the current smoking cessation market; and (2) a future market after the introduction of a new smoking cessation therapy (post-introduction).
- The model considered the following comparators: NicoDerm CQ, Nicotrol transdermal patch, generic transdermal patch, Nicorette, generic nicotine gum, Nicotrol Inhaler, Nicotrol NS, Commit Lozenge, Zyban, generic bupropion HCl, new prescription therapy, and no therapy.
- Input parameters for the model population and efficacy of comparators are displayed in Table 1.

Figure 1. Schematic of the Decision-Analytic Model

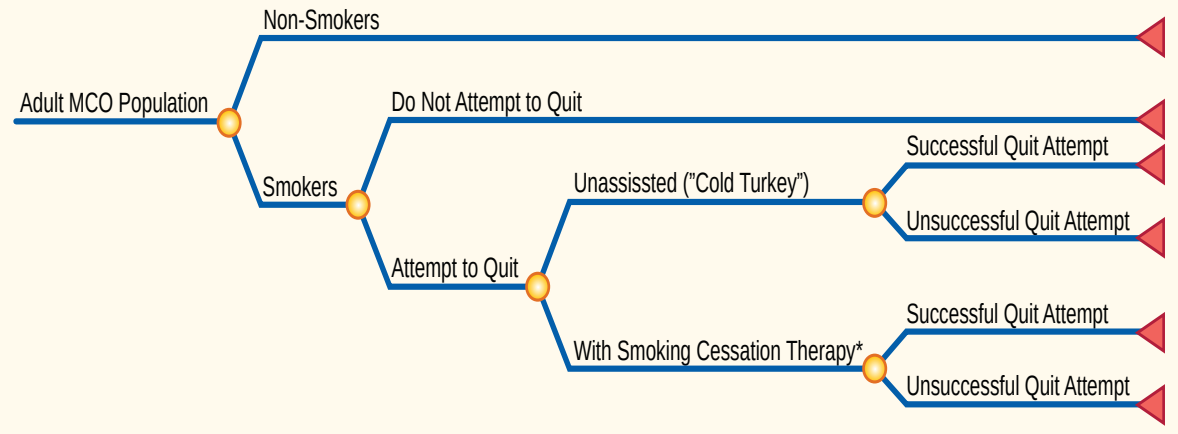

Adult MCO Population
Non-Smokers
Smokers
Do Not Attempt to Quit
Attempt to Quit
Unassissted ("Cold Turkey")
With Smoking Cessation Therapy*
Successful Quit Attempt
Unsuccessful Quit Attempt
Successful Quit Attempt
Unsuccessful Quit Attempt

Note: For simplification purposes, the "with smoking cessation therapy" arm is displayed as only one arm. In the model, this arm was made up of the various model comparators and thus was affected by the smoking cessation rates, drug and physician visit costs, and the market share of each comparator.

Table 1. Population and Efficacy Input Parameters and Sources

Population Input Parameter	Value
Percentage attempting to quit in the past year[11]	41.1%
Number of quit attempts per year*	2
Efficacy Input Parameter	
Unassisted ("cold turkey") quit rate[8, 9*]	4%
Commit lozenge[12*]	9%
Generic bupropion HCl[13]	9%
Generic nicotine gum[13]	8%
Generic nicotine transdermal patch[13]	6%
New prescription product[14*]	13%
NicoDerm CQ[13]	6%
Nicorette[13]	8%
Nicotrol inhaler[13]	8%
Nicotrol NS[13]	8%
Nicotrol transdermal patch[13]	6%
Zyban[13]	9%

*Assumption.

- The model assumed all therapies are dosed and used according to product labeling.
- Over-the-counter therapies were assumed to be not covered by the MCO.
- Prescription drugs were assumed to be placed on Tier 2 copayment levels with average US copayment costs.[15]
- One incremental physician visit was assumed to be needed for prescription therapy dosing or patient and/or adverse event monitoring. The cost of the visit is that of an established patient outpatient visit.[16]
- Drug costs were referenced to wholesale acquisition costs.[17]
- Assumed market share for the current market and post-introduction market are displayed in Table 2.[18]

Table 2. Product Mix Inputs

Comparator	Current Market	Post-Introduction Market
Unassisted ("cold turkey") quit rate	72.0%	70.6%
Committ Lozenge	3.8%	3.0%
Generic bupropion HCl	7.7%	6.2%
Generic nicotine gum	5.0%	4.0%
Generic nicotine transdermal patch	2.6%	2.1%
New prescription product	0.0%	7.0%
NicoDerm CQ	2.5%	2.0%
Nicorette	5.4%	4.3%
Nicotrol inhaler	0.7%	0.6%
Nicotrol NS	0.1%	0.1%
Nicotrol transdermal patch	0.1%	0.1%
Zyban	0.1%	0.1%

Source: Data on file; Assumption.

RESULTS

The prevalence of smoking by state and census region (Northeast, Midwest, South, and West) is presented in Figure 2.

Figure 2. Prevalence of Current Cigarette Smoking by Adults by State and Census Region

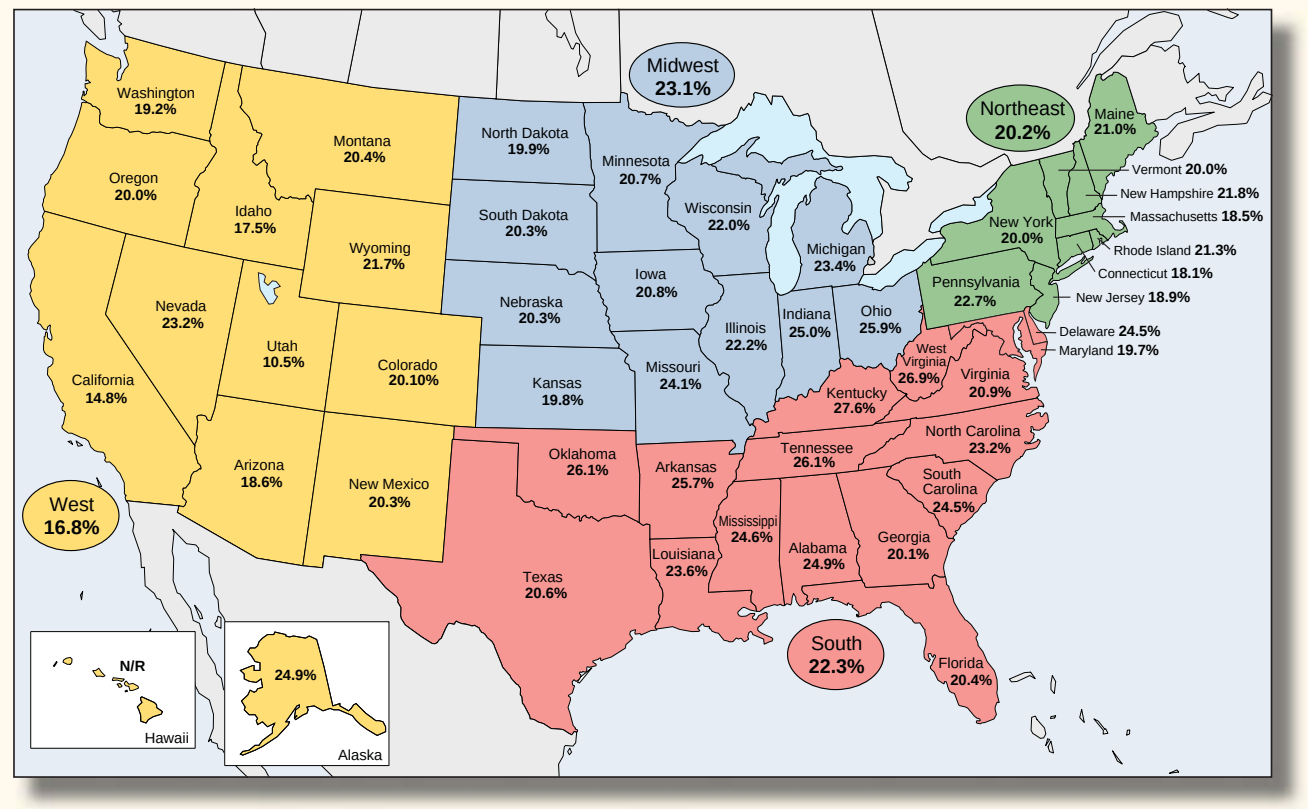

Washington 19.2%
Oregon 20.0%
Idaho 17.5%
Montana 20.4%
Wyoming 21.7%
Nevada 23.2%
Utah 10.5%
Colorado 20.10%
California 14.8%
Arizona 18.6%
New Mexico 20.3%
West 16.8%
N/R Hawaii
24.9% Alaska
Midwest 23.1%
North Dakota 19.9%
South Dakota 20.3%
Minnesota 20.7%
Wisconsin 22.0%
Iowa 20.8%
Nebraska 20.3%
Kansas 19.8%
Missouri 24.1%
Illinois 22.2%
Indiana 25.0%
Michigan 23.4%
Ohio 25.9%
Northeast 20.2%
Maine 21.0%
Vermont 20.0%
New Hampshire 21.8%
Massachusetts 18.5%
Rhode Island 21.3%
Connecticut 18.1%
New Jersey 18.9%
New York 20.0%
Pennsylvania 22.7%
Delaware 24.5%
Maryland 19.7%
West Virginia 26.9%
Virginia 20.9%
Kentucky 27.6%
Tennessee 26.1%
North Carolina 23.2%
South Carolina 24.5%
Oklahoma 26.1%
Arkansas 25.7%
Mississippi 24.6%
Louisiana 23.6%
Alabama 24.9%
Georgia 20.1%
Texas 20.6%
Florida 20.4%
South 22.3%

Source: CDC, 2005.
NR = not reported.

The annual per-member per-month (PMPM) cost for the current market and post-introduction market are presented in Figure 3.

Figure 3. Annual PMPM Cost by State and Census Region: Current Market/Post-Introduction Market

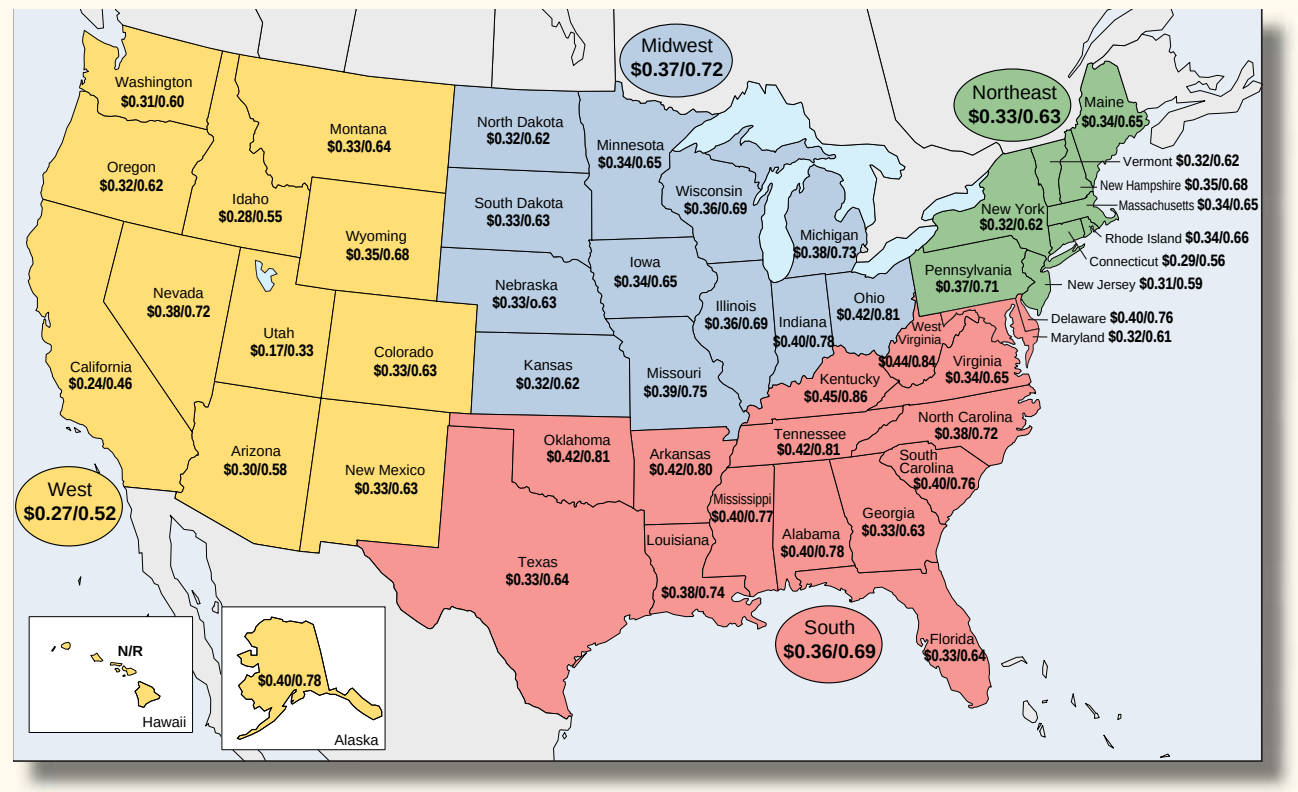

Washington $0.31/0.60
Oregon $0.32/0.62
Idaho $0.28/0.55
Montana $0.33/0.64
Wyoming $0.35/0.68
Nevada $0.38/0.72
Utah $0.17/0.33
Colorado $0.33/0.63
California $0.24/0.46
Arizona $0.30/0.58
New Mexico $0.33/0.63
West $0.27/0.52
N/R Hawaii
$0.40/0.78 Alaska
Midwest $0.37/0.72
North Dakota $0.32/0.62
South Dakota $0.33/0.63
Minnesota $0.34/0.65
Wisconsin $0.36/0.69
Iowa $0.34/0.65
Nebraska $0.33/0.63
Kansas $0.32/0.62
Missouri $0.39/0.75
Illinois $0.36/0.69
Indiana $0.40/0.78
Michigan $0.38/0.73
Ohio $0.42/0.81
Northeast $0.33/0.63
Maine $0.34/0.65
Vermont $0.32/0.62
New Hampshire $0.35/0.68
Massachusetts $0.34/0.65
Rhode Island $0.34/0.66
Connecticut $0.29/0.56
New Jersey $0.31/0.59
New York $0.32/0.62
Pennsylvania $0.37/0.71
Delaware $0.40/0.76
Maryland $0.32/0.61
West Virginia $0.44/0.84
Virginia $0.34/0.65
Kentucky $0.45/0.86
North Carolina $0.38/0.72
Tennessee $0.42/0.81
South Carolina $0.40/0.76
Oklahoma $0.42/0.81
Arkansas $0.42/0.80
Mississippi $0.40/0.77
Alabama $0.40/0.78
Georgia $0.33/0.63
Louisiana $0.38/0.74
Texas $0.33/0.64
Florida $0.33/0.64
South $0.36/0.69

NR = not reported.

Annual PMPM cost differences between the current market and post-introduction market are summarized in Figure 4.

Figure 4. Annual PMPM Cost Differences between the Current and Post-Introduction Markets

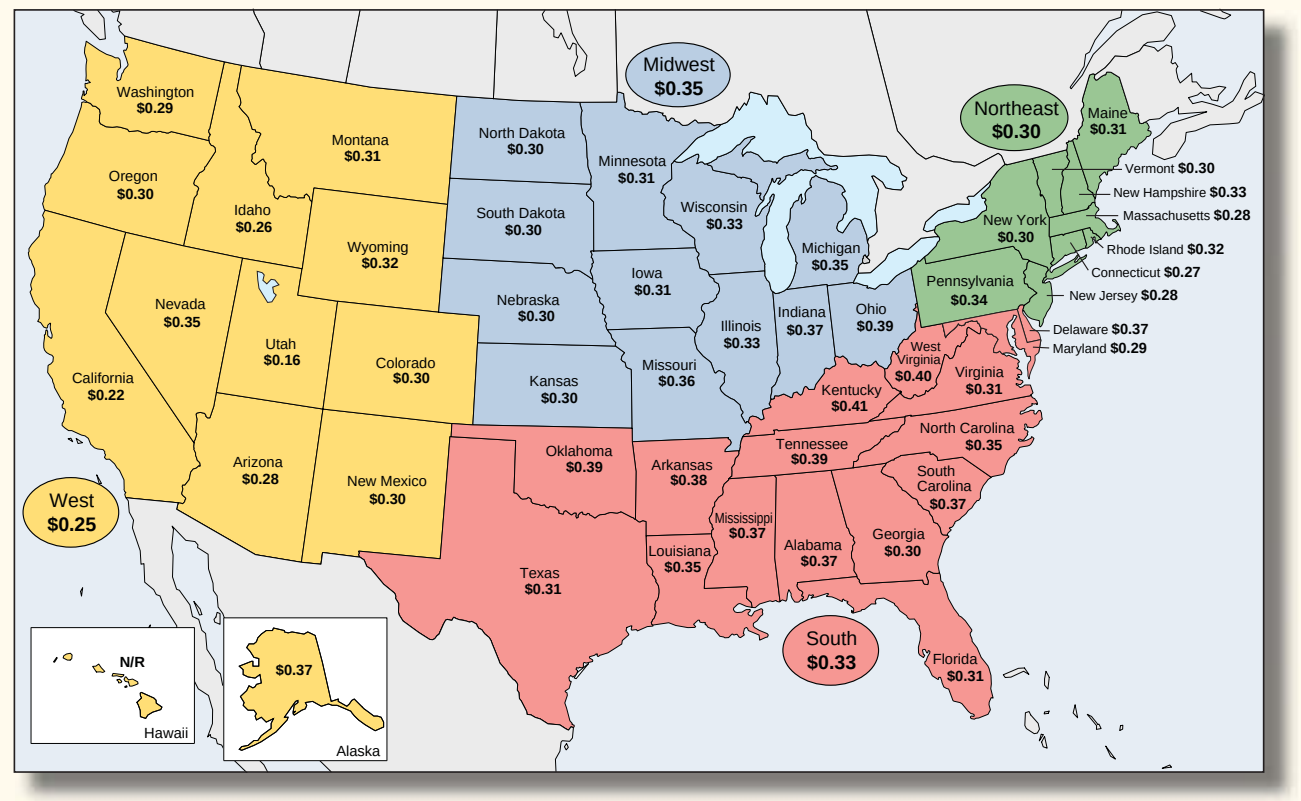

Washington $0.29
Oregon $0.30
Idaho $0.26
Montana $0.31
Wyoming $0.32
Nevada $0.35
Utah $0.16
Colorado $0.30
California $0.22
Arizona $0.28
New Mexico $0.30
West $0.25
N/R Hawaii
$0.37 Alaska
Midwest $0.35
North Dakota $0.30
South Dakota $0.30
Minnesota $0.31
Wisconsin $0.33
Iowa $0.31
Nebraska $0.30
Kansas $0.30
Missouri $0.36
Illinois $0.33
Indiana $0.37
Michigan $0.35
Ohio $0.39
Northeast $0.30
Maine $0.31
Vermont $0.30
New Hampshire $0.33
Massachusetts $0.28
Rhode Island $0.32
Connecticut $0.27
New Jersey $0.28
New York $0.30
Pennsylvania $0.34
Delaware $0.37
Maryland $0.29
West Virginia $0.40
Virginia $0.31
Kentucky $0.41
North Carolina $0.35
Tennessee $0.39
South Carolina $0.37
Oklahoma $0.39
Arkansas $0.38
Mississippi $0.37
Alabama $0.37
Georgia $0.30
Louisiana $0.35
Texas $0.31
Florida $0.31
South $0.33

NR = not reported.

- The introduction of a new smoking cessation therapy can increase annual PMPM cost by the following:
 - $0.16 to $0.41 when analyzed at the state-specific level, and
 - $0.25 to $0.35 when analyzed at the census region-specific level.

LIMITATIONS

- The model considered only physician visit and drug costs. The model did not consider the downstream cost benefits of quitting smoking. These costs could contribute to formulary decision making.
- Historical sales data were used to predict future outcomes that may be of interest to decision makers. The new product uptake will vary by region, managed care plan, and market.

CONCLUSIONS

- When modeling diseases or addiction behaviors with varying prevalence, it is important to analyze budgetary impact considering MCO population-specific prevalence values.
- Without micro-prevalence estimates, these costs could be underestimated or overestimated.
- The introduction of new smoking cessation therapies to the market will have a dramatic effect on an MCO's budget, regardless of the census region or state covered by the MCO. However, the impact can vary in magnitude.
- MCOs must use care in decision making in order to make accurate decisions on budgetary issues for new smoking cessation therapies.

REFERENCES

1. US Public Health Service. Treating tobacco use and dependence. 2000.
2. CDC. MMWR 2005a;54(20):509-12.
3. Bartecchi et al. N Engl J Med;330(13):907-12.
4. US Dept. of Health and Human Services. Reducing Tobacco Use: A Report of the Surgeon General. 2000.
5. Ockene et al. Health Psychol 1992;11(2):119-25.
6. Rosal et al. Health Psychol 1998;17(5):476-8.
7. Halpern et al. Tob Control 2001;10(3):233-8.
8. Hughes et al. Addiction 2004;99(1):29-38.
9. Cohen et al. Am Psychol 1989;44(11):1355-65.
10. Burton et al. MMWR 2000;49(29):665-8.
11. CDC. MMWR 2005b;54(44):1124-27.
12. Shiffman et al. Arch Intern Med 2002;162(11):1267-76.
13. West et al. Thorax 2000;5(12):987-99.
14. Gonzales et al. 12th Annual Meeting of the Society for Research on Nicotine and Tobacco. 2006 (abstract).
15. The Kaiser Family Foundation and Health Research and Education Trust Employer Health Benefits 2005 Annual Survey, 2005.
16. The Essential RBRVS, 2005.
17. Red Book for Windows. Thompson PDR 2006.
18. Data on File, GSK, 2006.